Final Results of a Randomized Phase 2 Study Showing the Clinical Benefit of Quizartinib (AC220) in Patients with FLT3-ITD Positive Relapsed or Refractory Acute Myeloid Leukemia

Gary Schiller, MD[1], Martin S Tallman, MD[2], Stuart Goldberg, MD[3], Alexander E Perl, MD[4], Jean-Pierre Marie, MD[5], Giovanni Martinelli, MD, PhD[6], Richard A Larson, MD[7], Nigel Russell, MD[8], Denise Trone[9], Guy Gammon, MD[9], Mark Levis, MD, PhD[10] and Jorge E. Cortes, MD[11]

[1]UCLA School of Medicine, Los Angeles, CA; [2]Leukemia Service, Department of Medicine, Memorial Sloan-Kettering Cancer Center, New York, NY; [3]Division of Leukemia, John Theurer Cancer Center at Hackensack University Medical Center, Hackensack, NJ; [4]Abramson Cancer Center of the University of Pennsylvania, Philadelphia, PA; [5]Department of Hematology, St-Antoine Hospital, Paris, France; [6]Institute of Hematology "L. e A. Seràgnoli", University of Bologna, Bologna, Italy; [7]University of Chicago, Chicago, IL, [8]University of Nottingham, Nottingham, UK, [9]Ambit Biosciences Corporation, San Diego, CA; [10]Sidney Kimmel Cancer Center at Johns Hopkins University, Baltimore, MD; [11]Department of Leukemia, The University of Texas, MD Anderson Cancer Center, Houston, TX.

385

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Background

- FLT3-ITD in AML is associated with early relapse and poor survival after treatment with standard chemotherapy
- Quizartinib is an orally active FLT3-ITD inhibitor
- A previous Phase 2 study included subjects with FLT3-ITD(+) AML, relapsed/refractory to 2nd line chemotherapy or relapsed after hematopoietic stem cell transplant (HSCT), doses of 90, 135 or 200 mg/day
 - 46% of subjects achieved CRc (CRc = CR + CRp + CRi)
 - 35% of subjects were bridged to HSCT (mostly after achieving CRi)
 - 33% of subjects with a CRi and bridged to HSCT were alive beyond 1 year
 - Quizartinib was generally well tolerated
 - Grade 3 QTcF was observed in 18% of subjects
 - No Grade 4 QTcF prolongation was recorded

Study Design and Population

- This was an open label, randomized comparison of two doses of quizartinib, 30 mg/day and 60 mg/day, given for 28 day continuous cycles, to FLT3-ITD(+) subjects who were ≥18yrs and were relapsed or refractory to salvage therapy or relapsed after HSCT
- Subjects had their dose reduced for any of the following reasons:
 - Grade 2 or higher QTcF prolongation
 - Grade 3 or higher related non-hematologic toxicity
 - Myelosuppression for subjects in CRc
- Subjects could have their dose increased for lack of CRc after 1 cycle or loss of response
- The primary endpoints were:
 - CRc Rate (CRc = CR + CRp + CRi)
 - Incidence of ≥ Grade 2 QTcF Prolongation
- The secondary endpoints included:
 - Overall Survival
 - Duration of Response
 - Bridge to Transplant Rate
 - Safety
- Analysis based on data with a cutoff taken at 52 weeks following randomization of the last subject into the study
- IWG response criteria for CRi and PR were modified and subjects did not have to be transfusion independent. CR and CRp were according to IWG Criteria

Table 1. Patient Characteristics

	N (%) or Median [range]		
	30 mg/day (N=38)	60 mg/day (N=38)	Total (N=76)
Age (years)	57 [19-77]	53 [20-74]	55 [19-77]
Age ≥60 years	16 (42)	10 (26)	26 (35)
Males	22 (58)	22 (58)	44 (58)
ECOG PS 2	7 (18)	5 (14)	12 (16)
Secondary AML	3 (8)	7 (18)	10 (13)
FLT3-ITD(+) allelic ratio	35 (92)	35 (92)	70 (92)
>10% and ≤25%	9 (24)	5 (13)	14 (18)
≥25% and ≤50%	20 (53)	13 (34)	33 (43)
>50%	6 (16)	17 (45)	23 (30)
Intermediate Cytogenetic Risk*	26 (68)	25 (66)	51 (67)
Poor Cytogenetic Risk*	4 (11)	3 (8)	7 (9)

*Cytogenetic information based on available data

Table 2. Prior Treatment and Response

	30 mg/day (N=38)	60 mg/day (N=38)	Total (N=76)
Refractory N (%)	9 (24)	13 (34)	22 (29)
Relapsed N (%)	29 (76)	24 (63)	53 (70)
CR1 (weeks) Median [range]	17.4 [2.1, 60.9]	26.1 [3.6, 78.3]	26.1
Previous HSCT N (%)	9 (24)	12 (32)	21 (28)
Median time from previous HSCT (weeks)	23.6	28.1	-
Prior FLT3 Therapy N (%)	5 (13)	7 (18)	12 (16)
Median time from end of prior FLT3 therapy to start of quizartinib treatment (weeks)	4.9	13.3	-

All 12 subjects taking a prior FLT3 therapy received sorafenib. 1 also received midostaurin and 1 also received crenolanib

Table 3. Patient Disposition

	30 mg/day (N=38)	60 mg/day (N=38)	Total (N=76)
Active treatment	0	1 (3)	1 (1)
Discontinued	38 (100)	37 (97)	75 (99)
Relapse / Lack of Efficacy	19 (50)	16 (42)	35 (46)
HSCT	11 (29)	15 (39)	26 (34)
Adverse event(s)*	6 (16)	1 (3)	7 (9)
Subject withdrawal	1 (3)	2 (5)	3 (4)
Randomized no tx	0	2 (5)	2 (3)
Death**	1 (3)	1 (3)	2 (3)

*3 of 7 AEs leading to discontinuation were related to study drug and all were in the 30 mg/day group: neutropenic sepsis, diarrhea, and pleural effusion. The last 2 events occurred after the subject had escalated to 60 mg/day
**2 deaths, one due to AML progression and one due to sepsis, disseminated intravascular coagulation + multi-organ failure – both considered unrelated to quizartinib

Table 4. Quizartinib Treatment Duration and Dose Modifications*

	Median [range] or N (%)		
	30 mg/day (N=38)	60 mg/day (N=36**)	Total (N=74)
Median Duration of Treatment (range), weeks	9.4 [2.1, 33.4]	10.2 [2.6, 52+]	
Dose Interrupted	14 (37)	17 (47)	31 (42)
Dose Reduced	12 (32)	11 (31)	23 (31)
QTc prolongation	1	2	3
Other adverse events	2	2	4
Myelosuppression	4	6	10
Dose Escalated	24 (63)	7 (19)	31 (42)
No response	11	2	13
Loss of response	13	5	18

*At the time of last dose, 5 patients were receiving 20 mg/day, 22 patients were receiving 30 mg/day, 42 patients were receiving 60 mg/day and 5 patients were receiving 90 mg/day
**Two patients were randomized but did not receive drug due to ineligibility

Table 5. Treatment-Emergent AEs: Incidence ≥25%

	N (%)			
	30 mg/day (N=38)		60 mg/day (N=36)	
Adverse Event	All grades	Grade 3/4	All grades	Grade 3/4
Any event	37 (97)	30 (79)	36 (100)	32 (89)
Anemia	18 (47)	14 (37)	9 (25)	6 (17)
Fatigue	13 (34)	1 (3)	8 (22)	2 (6)
Pyrexia	11 (29)	3 (8)	13 (36)	3 (8)
Vomiting	11 (29)	0	13 (36)	3 (8)
Febrile neutropenia	11 (29)	11 (29)	13 (36)	13 (36)
Diarrhea	10 (26)	1 (3)	13 (36)	1 (3)
Cough	9 (24)	1 (3)	9 (25)	0
Nausea	9 (24)	0	17 (47)	3 (8)
Abdominal Pain	6 (16)	0	11 (31)	0
Headache	4 (11)	1 (3)	9 (25)	1 (3)

Table 6. Overall Response

	N (%)		
Best Response	30 mg/day* (N=38)	60 mg/day* (N=38)	Total (N=76)
CRc (CR+CRp+CRi)	18 (47)	18 (47)	36 (47)
CR	2 (5)	1 (3)	3 (4)
CRp	0	1 (3)	1 (1)
CRi	16 (42)	16 (42)	32 (42)
PR	5 (13)	9 (24)	14 (18)
Duration of CRc* weeks [95% CI]	4.1 [3.9, 9.7]	20.0 [4.3, 20]	5.3 [4.1, 12.3]

*30 mg/day arm: 7/18 censored; 5 censored due to HSCT
60 mg/day arm: 11/18 censored; 9 censored due to HSCT

Figure 1. Overall Survival by Dose

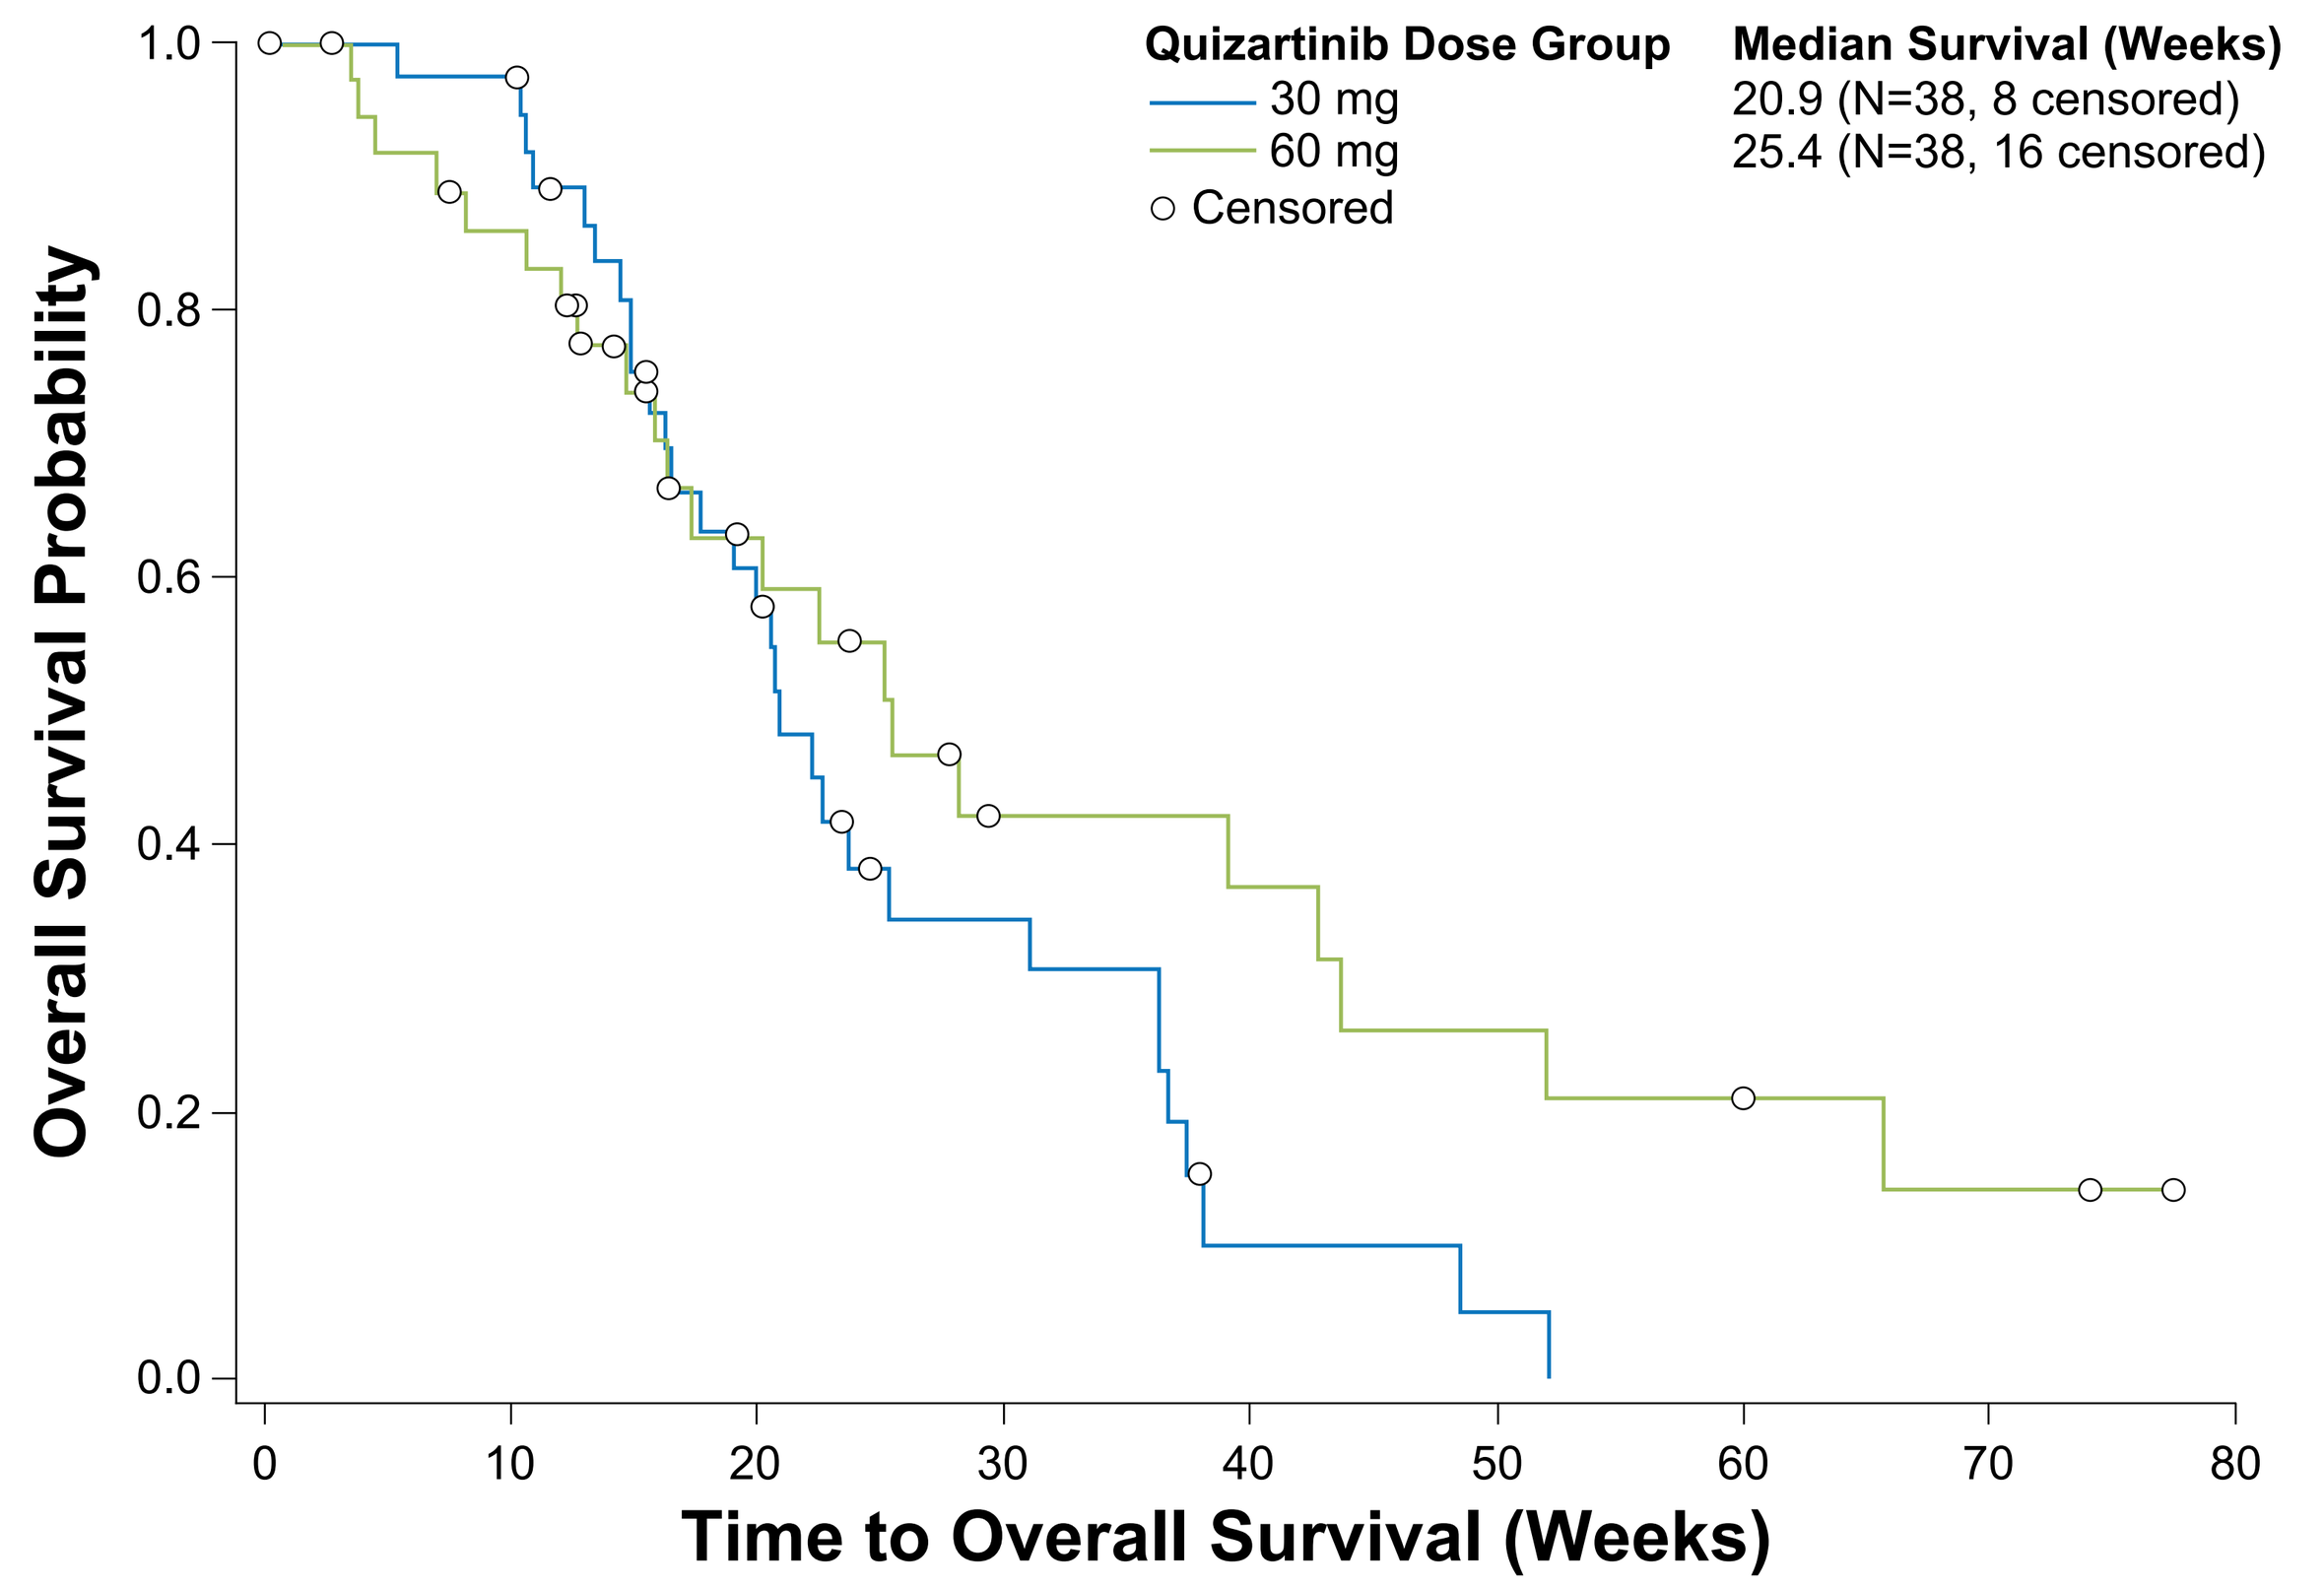
Quizartinib Dose Group
30 mg
60 mg
Censored
Median Survival (Weeks)
20.9 (N=38, 8 censored)
25.4 (N=38, 16 censored)
Overall Survival Probability
Time to Overall Survival (Weeks)

Figure 2. Overall Survival by Dose and HSCT Post-Quizartinib

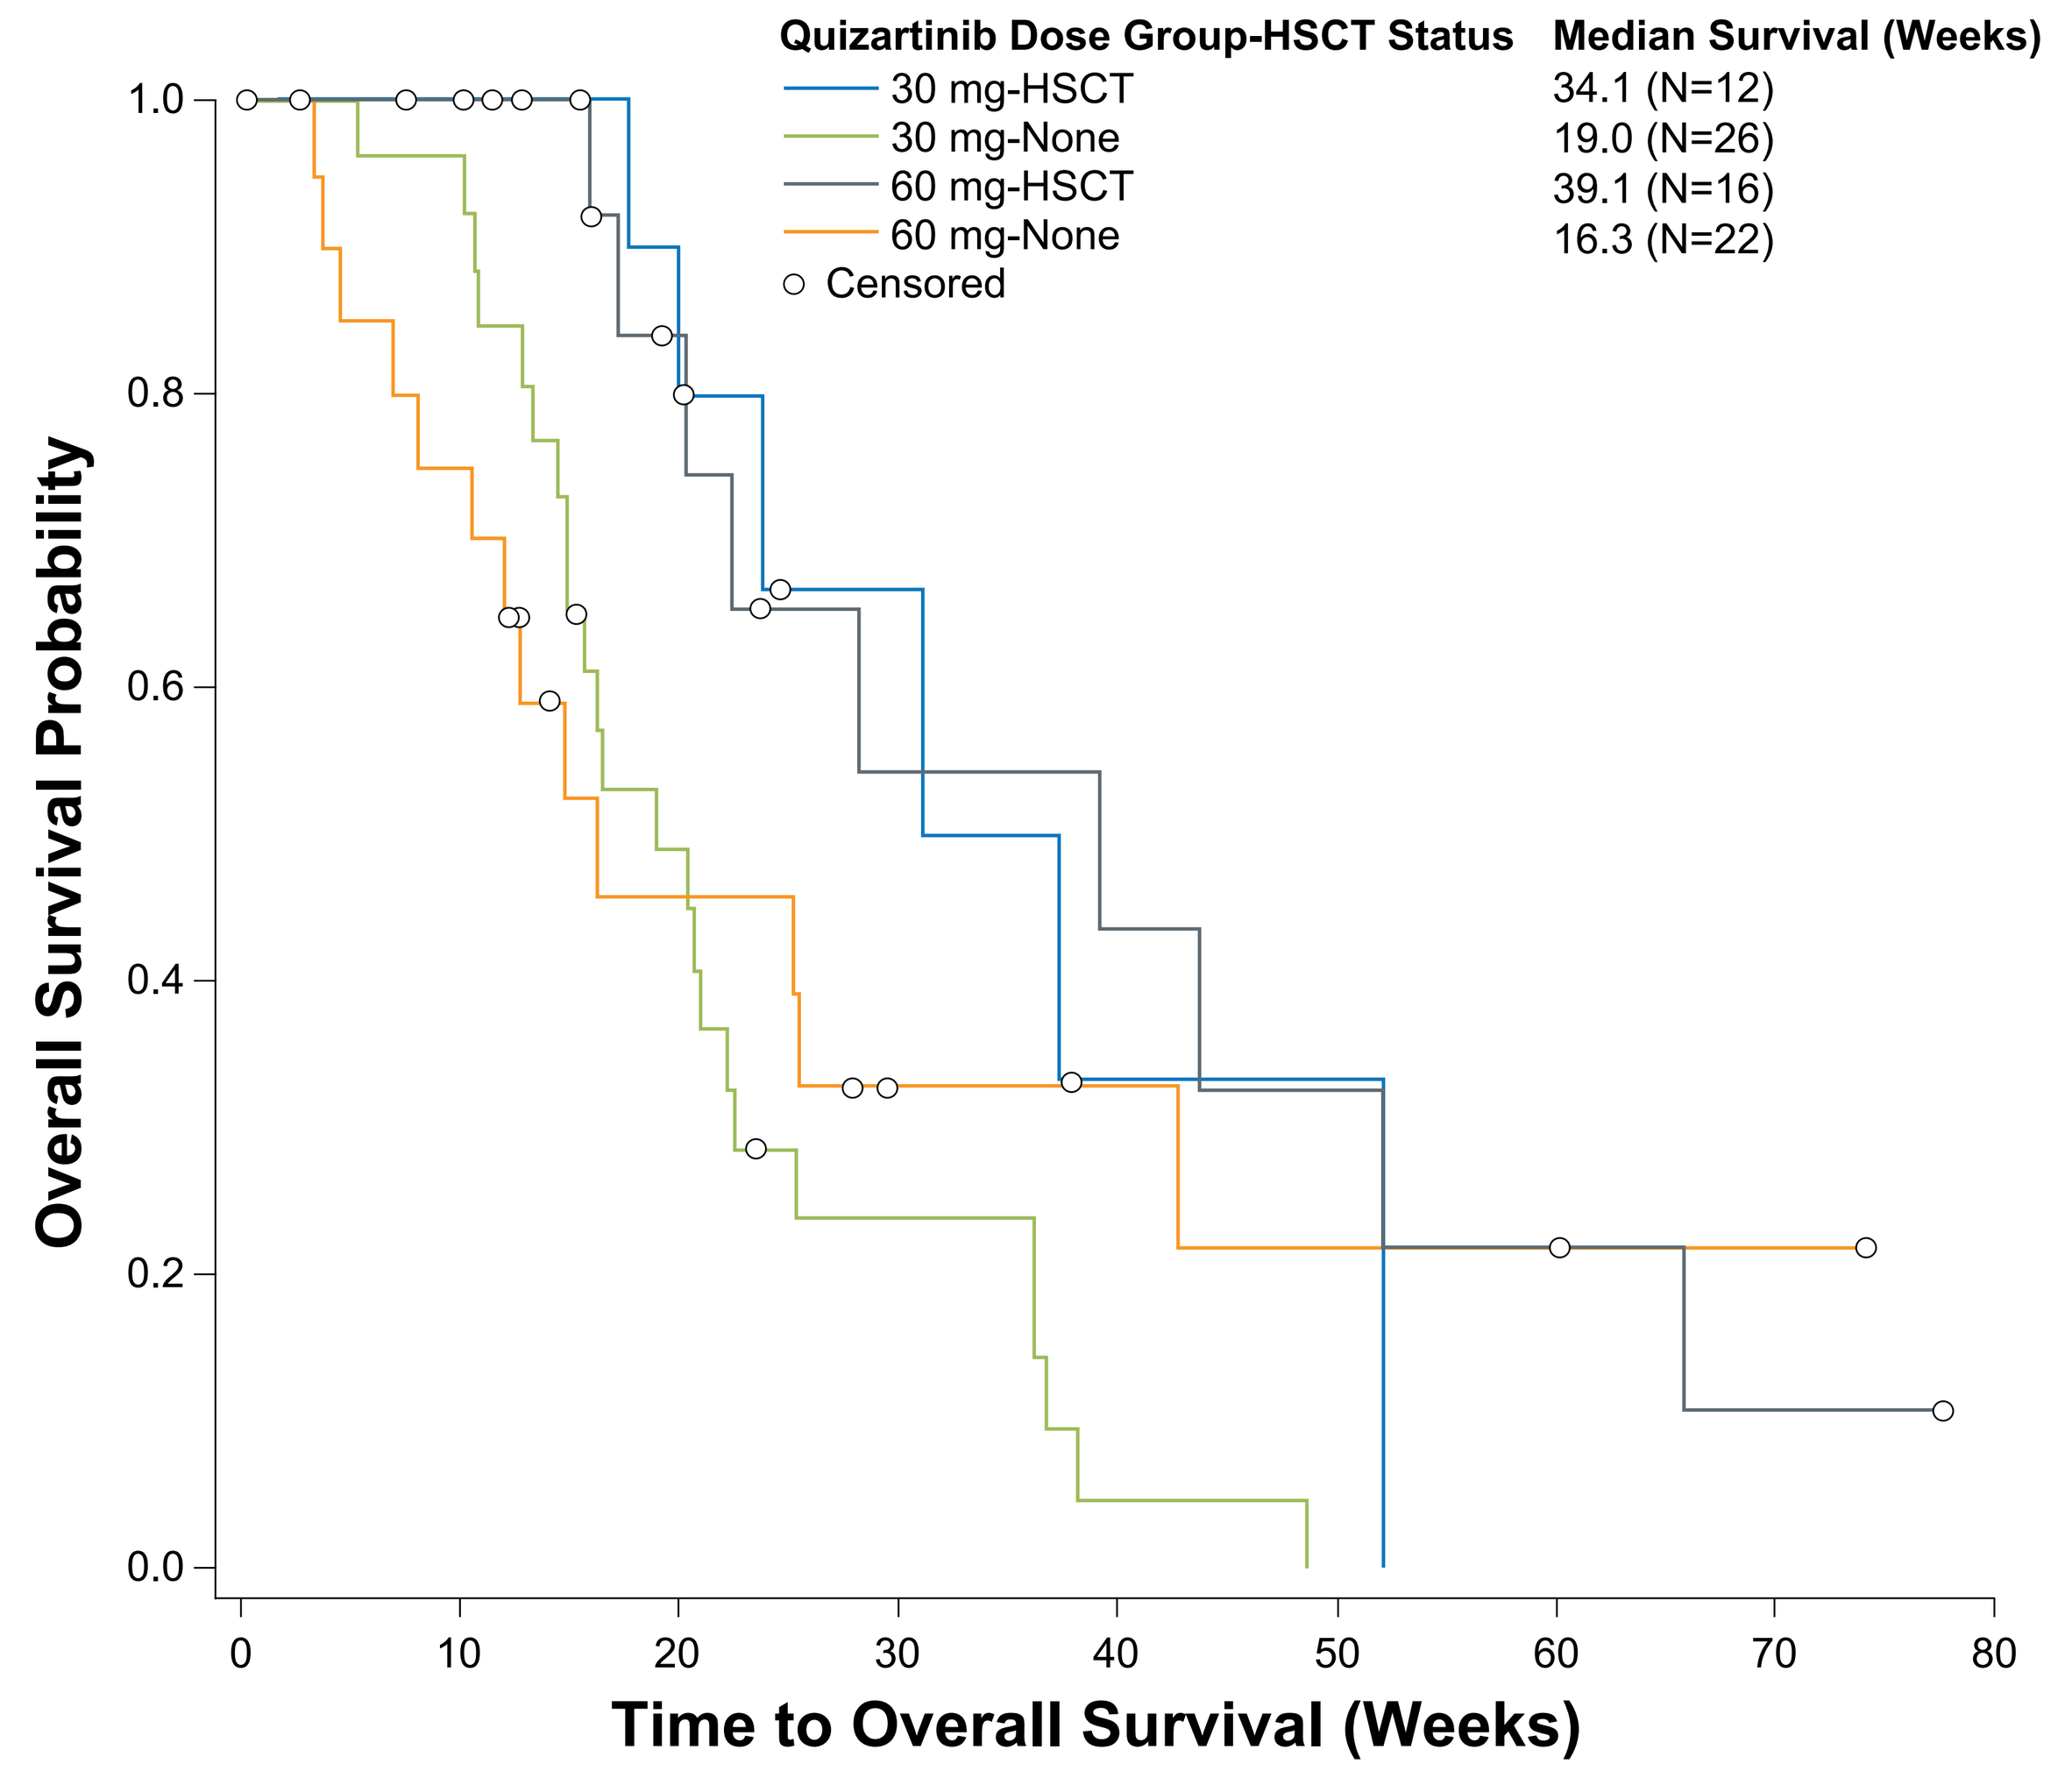
Quizartinib Dose Group-HSCT Status
30 mg-HSCT
30 mg-None
60 mg-HSCT
60 mg-None
Censored
Median Survival (Weeks)
34.1 (N=12)
19.0 (N=26)
39.1 (N=16)
16.3 (N=22)
Overall Survival Probability
Time to Overall Survival (Weeks)

Table 7. Updated Results: Bridge to Transplant

	N (%)	
	30 mg/day (N=38)	60 mg/day (N=38)
Bridge to Transplant Rate	12 (32)	16 (42)*
CR	2 (5)	1 (3)
CRi	7 (18)	9 (24)
PR	1 (3)	3 (8)
NR	2 (5)	3 (8)
Median Survival (weeks)	34.1	39.1
Alive	6 (50)	7 (44)
Died	6 (50)	9 (56)

*2 subjects went to HSCT but listed progressive disease and adverse event as the reasons for treatment discontinuation

Long-term Survivors

- 24 subjects were alive at >24 weeks and 35 had died (17 were censored before 24 weeks)
- 4 subjects remained alive >12 months and 51 had died (21 censored before 1 year follow up). These 4 subjects were classified as "long-term survivors"
 - 3/4 long-term survivors achieved at least a PR (2 CRi and 1 PR) to quizartinib
 - 2/4 (50%) long-term survivors went to HSCT after receiving quizartinib and were on quizartinib for 10 and 14.6 weeks
 - 1/4 (25%) long-term survivors remain on study taking quizartinib for over 1 year

Table 8. Comparison of Quizartinib Doses in Two Phase 2 Studies in >200 Subjects

	2689-CL-2004		AC220-002 (Cohort 2)		
	30 mg/day (N=38)	60 mg/day (N=38)	90 mg/day (N=57)	135 mg/day (N=67)	200 mg/day (N=12)
Best Response					
CRc Rate	47%	47%	47%	45%	42%
PR Rate	13%	24%	25%	28%	50%
Maximum value of QTcF (msec)					
>450 to ≤480	42%	47%	35%	48%	25%
>480 to ≤500	5%	14%	21%	13%	33%
>500	5%	3%	21%	15%	42%
Maximum change in QTcF from baseline (msec)					
≤30	50%	44%	9%	9%	0%
>30 to ≤60	47%	36%	46%	51%	8%
>60	3%	19%	46%	39%	92%

Summary

- Quizartinib at doses of 30 and 60 mg daily, in second salvage or post HSCT in FLT3-ITD(+) AML subjects, demonstrated a high degree of activity (CRc 47%) equivalent to higher daily doses (90, 135 and 200 mg) administered in the previous Phase 2 study
- Bridge to transplant rate was equivalent to that at higher doses; improved median survival for HSCT subjects in updated data
- Overall survival was equivalent to previous Phase 2 study at higher doses
- There was a decrease in QTcF prolongation at lower doses together with an acceptable safety profile

Further Studies

- A Phase 3 study comparing the effect on overall survival of either quizartinib (starting dose of 30 mg/day) or standard chemotherapy in subjects with relapsed/refractory FLT3-ITD AML is underway

Acknowledgments

We would like to thank all participating patients and their families, and the network of investigators, research nurses, study coordinators, and operations staff.

- Study 2689-CL-2004 (ClinicalTrials.gov identifier NCT01565668) was supported by Ambit Biosciences Corporation.

Study Investigators:

- UNITED STATES: J. Altman, M. Baer, D. Claxton, J. Cortes, S. Goldberg, R. Larson, M. Levis, M. Litzow, A. Perl, G. Schiller, S. Strickland, R. Stuart, M. Tallman, C. Ustun
- EUROPE: C-E. Bulabois, M. Hunault-Berger, J-P. Marie, G. Martinelli, A. Pigneux, B. Quesnel, N. Russell, N. Vey

The 50th Annual Meeting of the American Society of Clinical Oncology; May 30–June 3, 2014; Chicago, Illinois